Clinical Remission with Beclometasone/Formoterol/Glycopyrronium in Asthma: Real-World Evidence from the TriMaximize Study

C. Gessner[1], F. Trinkmann[2], V. Bogoevska[3], REK. Russell[4], C. Suppli Ulrik[5], W. Pohl[6], V. Plaza[7], A. Bourdin[8], M. Kupczyk[9], F. Braido[10], C. Fritz[3], A. Hahn[3], V. Grickschat[3], M. Lommatzsch[11].

[1]Specialized Practice for Pulmonary Medicine Leipzig, Leipzig, [2]Department of Pneumology and Critical Care Medicine, Thoracic clinic, University of Heidelberg, Heidelberg, [3]Cheisi GmbH, Hamburg, [4]King's College London, London, [5]Department of Respiratory Medicine, Copenhagen University Hospital-Hvidovre, Hvidovre, [6]Karl Landsteiner Institute for Clinical and Experimental Pneumology, Clinic Hietzing, Vienna, [7]Hospital de la Santa Creu i Sant Pau, Barcelona, [8]Hôpital Arnaud de Villeneuve, University of Montpellier, Montpellier, [9]Department of Internal Medicine, Asthma and Allergy, Medical University of Lodz, Lodz, [10]Università degli Studi di Genova - Ospedale San Martino, Genova , [11]Department of Internal Medicine, University of Rostock, Rostock.

Scan Me

TR:MAXIMIZE

BACKGROUND:

- Randomized clinical trials have shown clinical efficacy of extrafine formulation single-inhaler triple therapy consisting of beclometasone dipropionate/formoterol fumarate/glycopyrronium (BDP/FF/G)[1].
- TriMaximize study observes patients who have switched to BDP/FF/G in a real-world setting over a period of one to three years[2].

METHODS:

- TriMaximize is a multinational, observational study that follows patients with asthma who have been prescribed BDP/FF/G up to 36 months. Patients were recruited at 162 sites across eight countries (Germany, United Kingdom, Austria, Denmark, France, Spain, Poland and Italy). Study-typical attrition resulted in a visit-dependent decline in patient numbers (baseline n=1445; 3 months n=919; 12 months n=800; 24 months n=281; 36 months n=63), due to routine drop-outs and missing follow-up data.
- Two-component clinical remission was defined by an absence of exacerbations and systemic corticosteroid use for at least one year, three-component remission was defined by an additional Asthma Control Test (ACT) score ≥ 20 and four-component remission was defined by an additional lung function criterion. Four different lung function criteria for 4-components analysis were explored: (1) FEV_1 decline ≤ 5% predicted, (2) FEV_1 ≥ 80% predicted, (3) FEV_1 decline ≤ 5% predicted or FEV_1 ≥ 80% predicted, (4) improvement in FEV_1 by ≥ 100 mL. Patients with intake of biologics during the follow-up period were excluded.

Table 1. Baseline characteristics.

Parameters		Overall population n= 1445	Medium strength (87/5/9 µg) n= 1084	High strength (172/5/9 µg) n= 361
Age (years), mean (±SD)		57.6 (15.0)	57.9 (14.8)	56.4 (15.4)
Sex, n (%)	Female	907 (62.8)	685 (63.2)	222 (61.5)
	Male	538 (37.2)	399 (36.8)	139 (38.5)
BMI (kg/m^2), mean (±SD)		28.9 (6.4)	28.8 (6.4)	29.2 (6.5)
Smoking status, n (%)	Never smoker	755 (52.2)	561 (51.8)	194 (53.7)
	Current smoker	259 (17.9)	203 (18.7)	56 (15.5)
	Former smoker	431 (29.8)	320 (29.5)	111 (30.7)
Time since stopped smoking (years)* (±SD)		14.7 (12.4)	14.1 (12.2)	16.5 (13.1)
Time since diagnosis at baseline visit, years (±SD)		15.1 (14.7)	13.6 (13.6)	19.7 (16.8)
Concomitant diseases, n (%)		1161 (85.3)	847 (84.1)	314 (88.7)
	Arterial hypertension	507 (37.8)	392 (39.2)	115 (33.7)
	COPD	298 (22.4)	255 (25.7)	43 (12.8)
Rate of moderate or severe asthma exacerbations in previous year, mean (±SD)		1.8 (1.6)	1.8 (1.5)	2.0 (1.9)
GINA classification, n (%)	GINA Step 4	1079 (76.3)	913 (85.3)	166 (48.3)
	GINA Step 5	335 (23.7)	157 (14.7)	178 (51.7)
Prior treatment	ICS/LABA**	1050 (72.7)	854 (78.8)	196 (54.3)
	ICS/LABA/LAMA**	395 (27.3)	230 (21.2)	165 (45.7)

*restricted to former smokers
**fixed or free

Presented at ATS International Conference 2026, Orlando May 15-20

RESULTS:

Figure 1. Proportion of patients achieving clinical remission components.

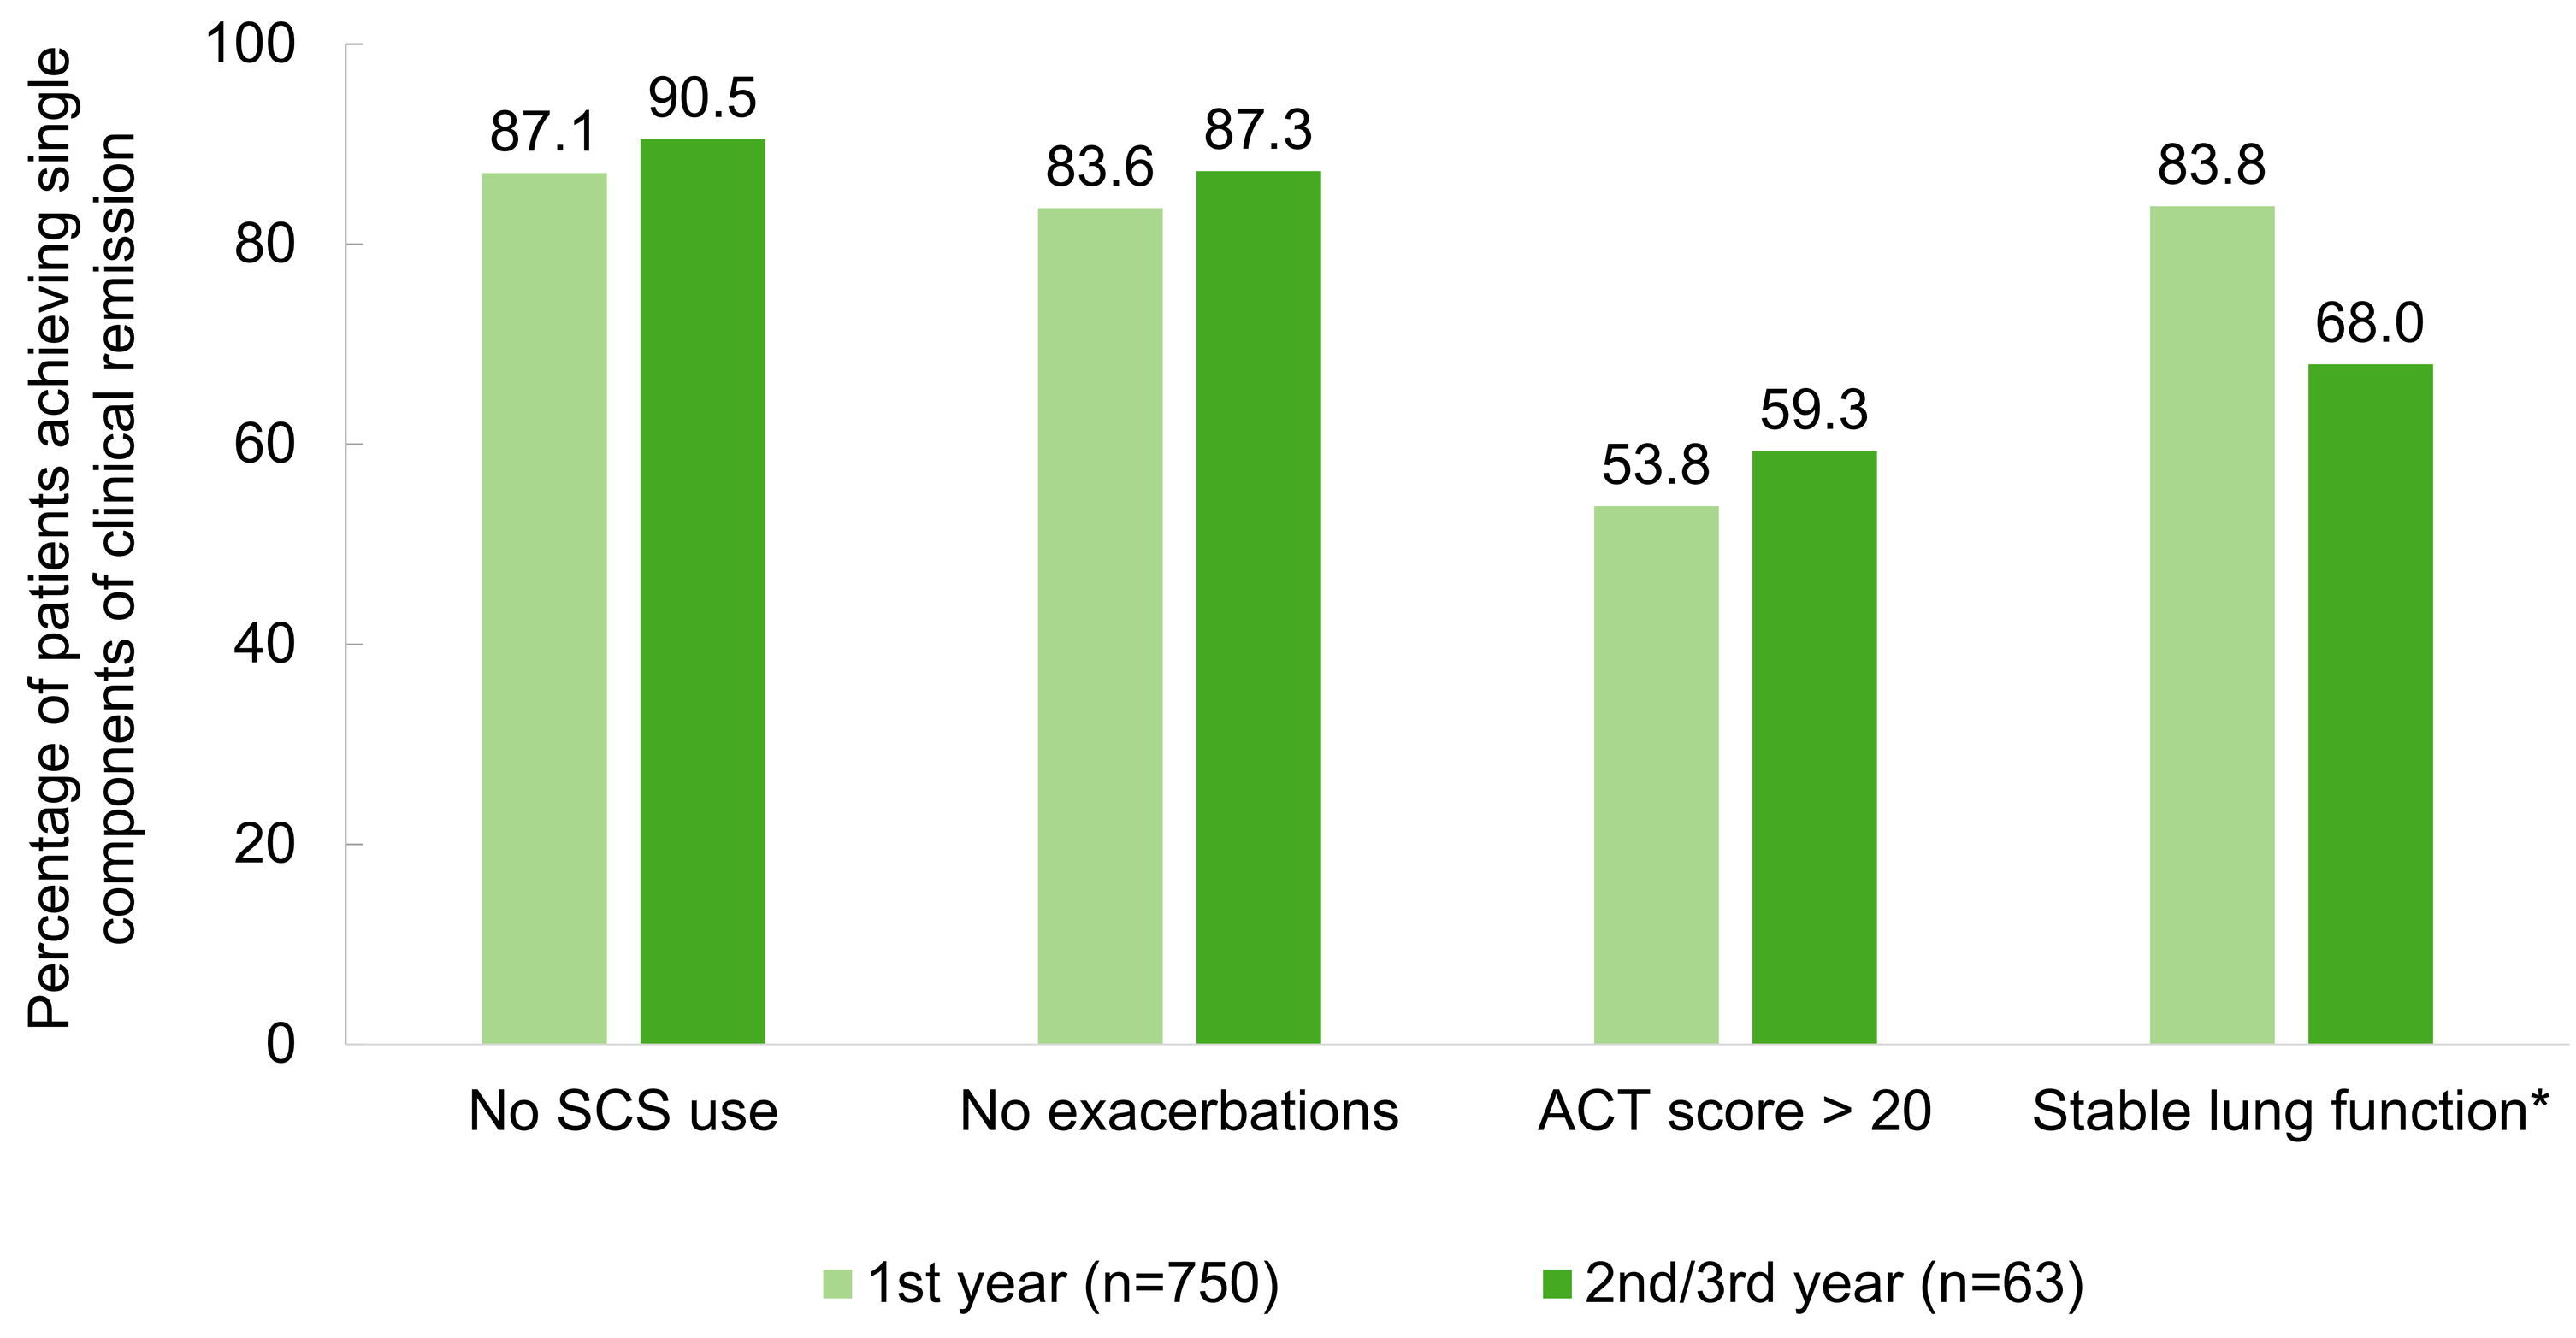
Percentage of patients achieving single components of clinical remission
100
80
60
40
20
0
87.1
90.5
83.6
87.3
53.8
59.3
83.8
68.0
No SCS use
No exacerbations
ACT score > 20
Stable lung function*
1st year (n=750)
2nd/3rd year (n=63)

*Stable lung function: FEV_1 improvement or decline of no more than 5% of predicted (FEV_1 decline ≤ 5%)
FEV_1 - forced expiratory volume in 1 second

Figure 2. Clinical remission (2 components, 3 components, 4 components).

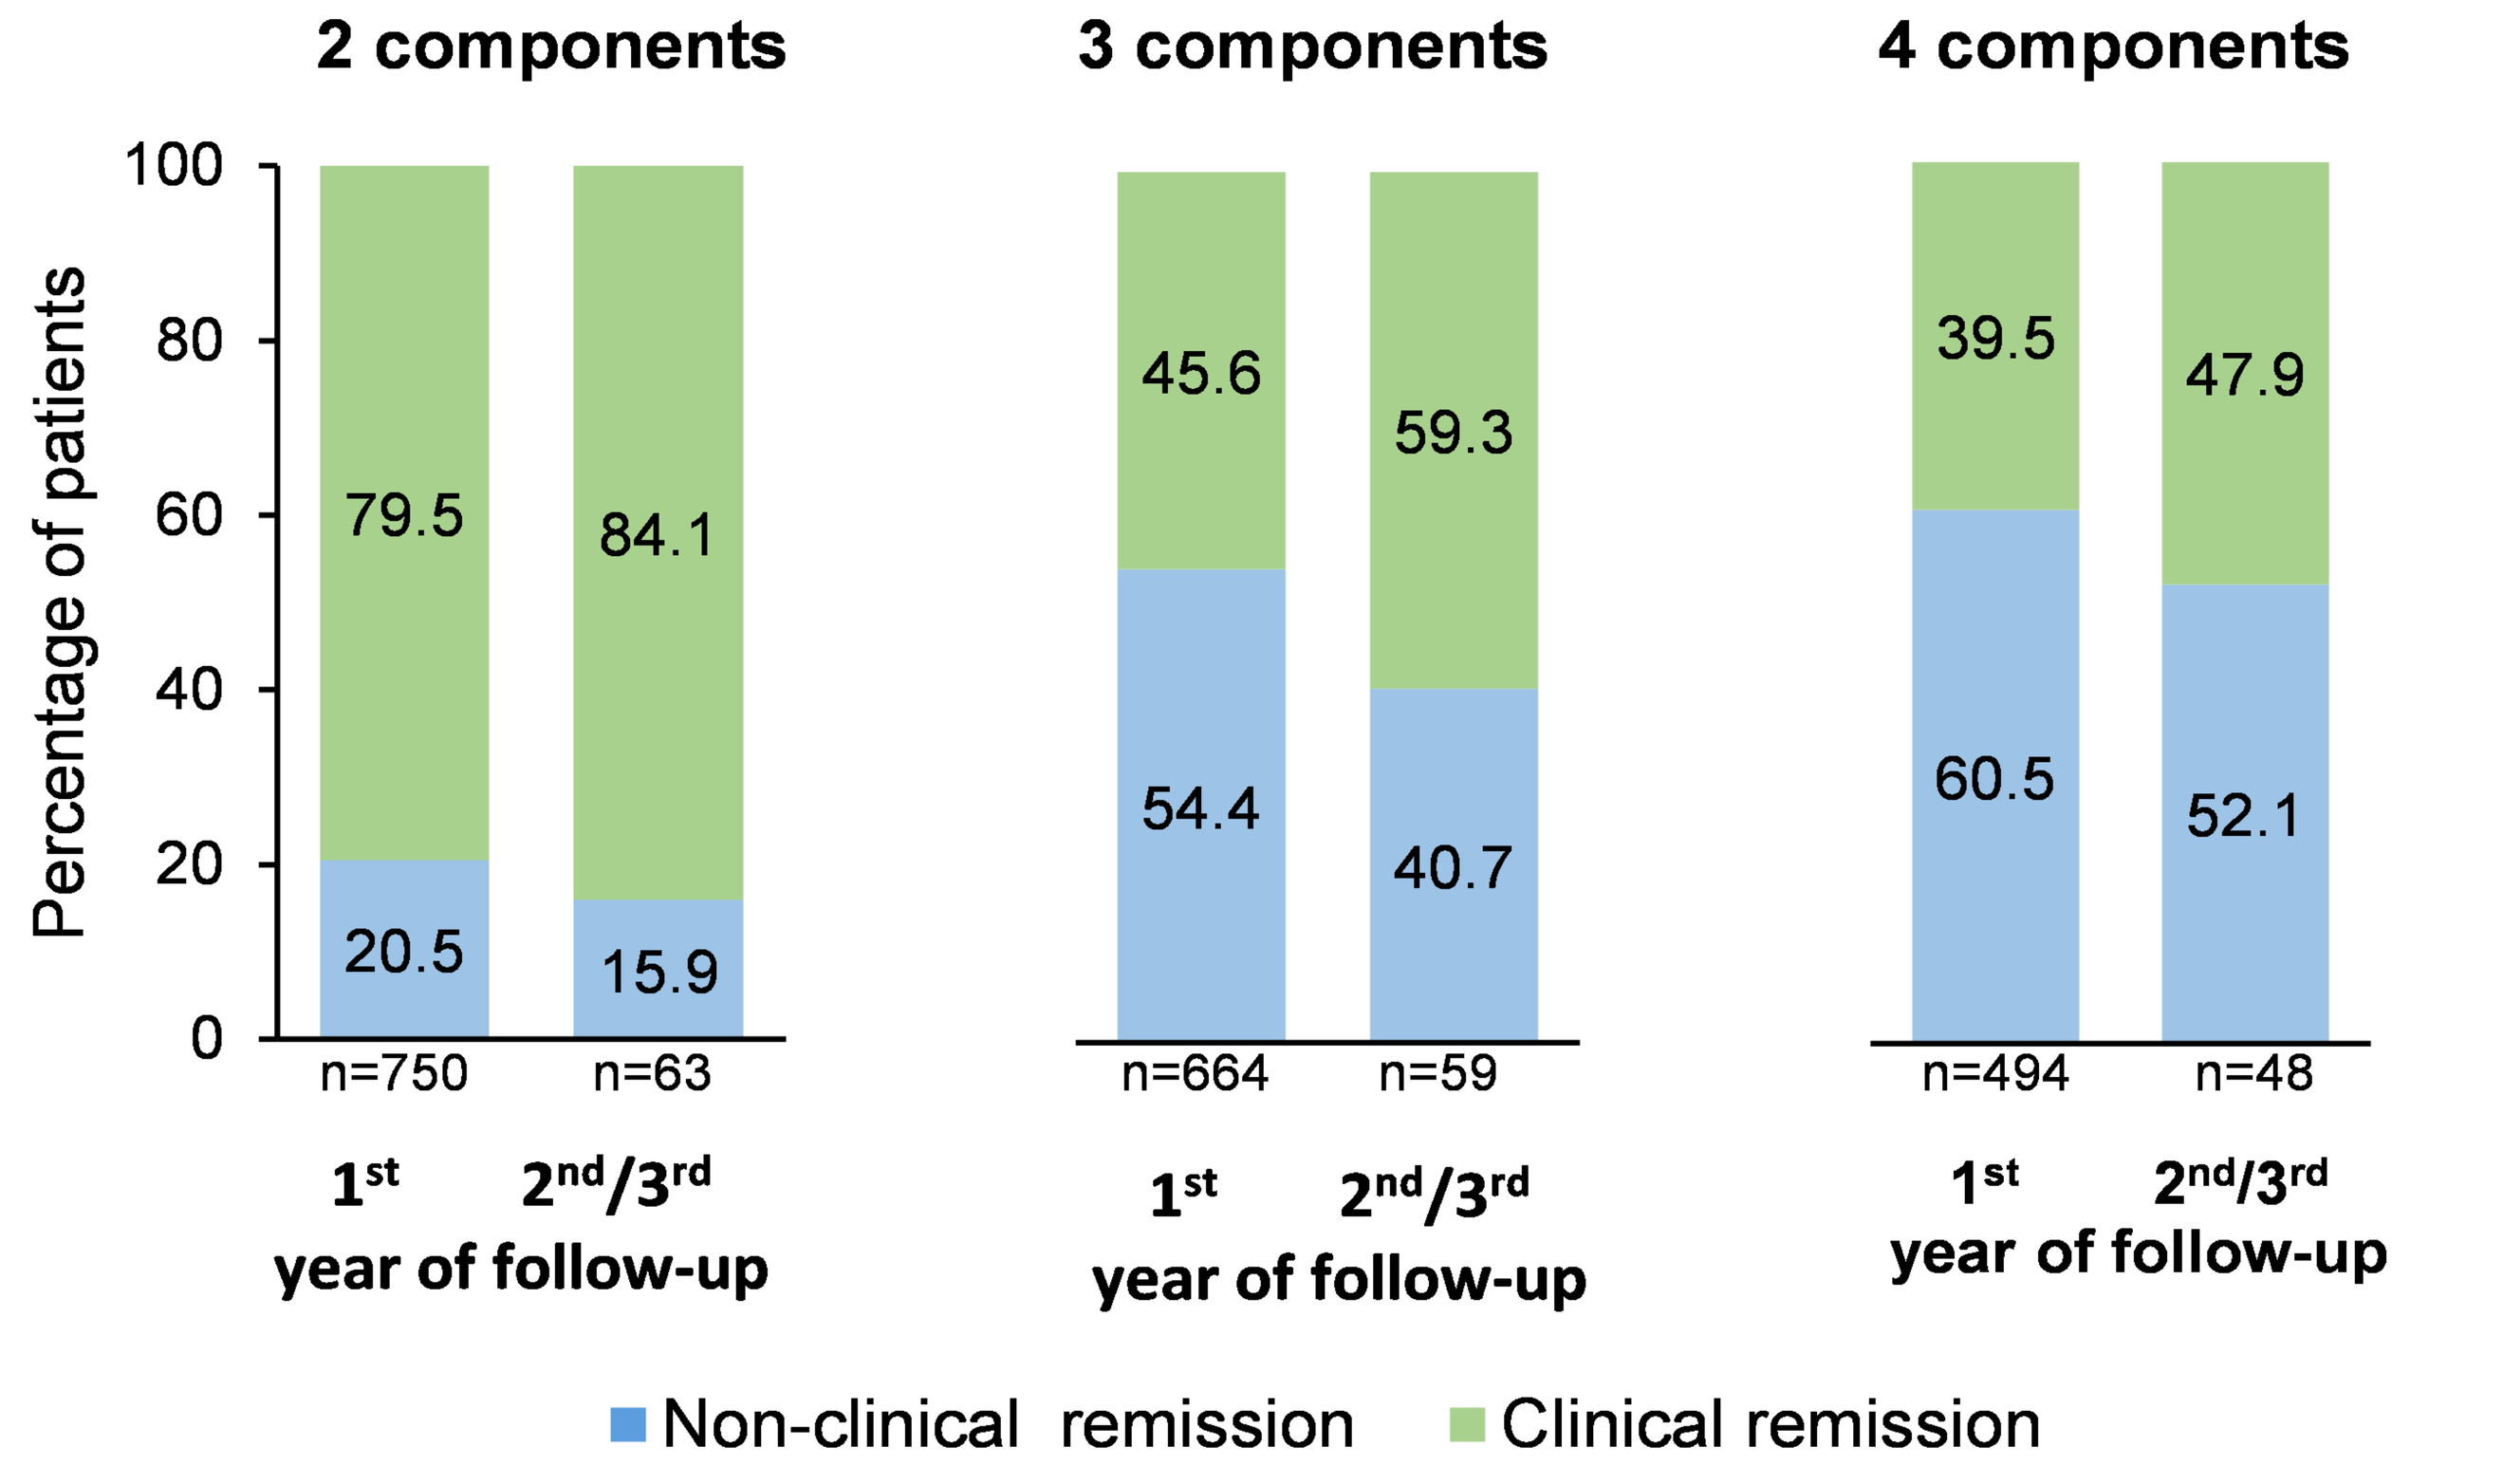
2 components
3 components
4 components
Percentage of patients
100
80
60
40
20
0
79.5
84.1
20.5
15.9
45.6
59.3
54.4
40.7
39.5
47.9
60.5
52.1
n=750
n=63
n=664
n=59
n=494
n=48
1st 2nd/3rd
year of follow-up
Non-clinical remission
Clinical remission

Figure 3. Clinical remission (4 components with different FEV_1 criteria).

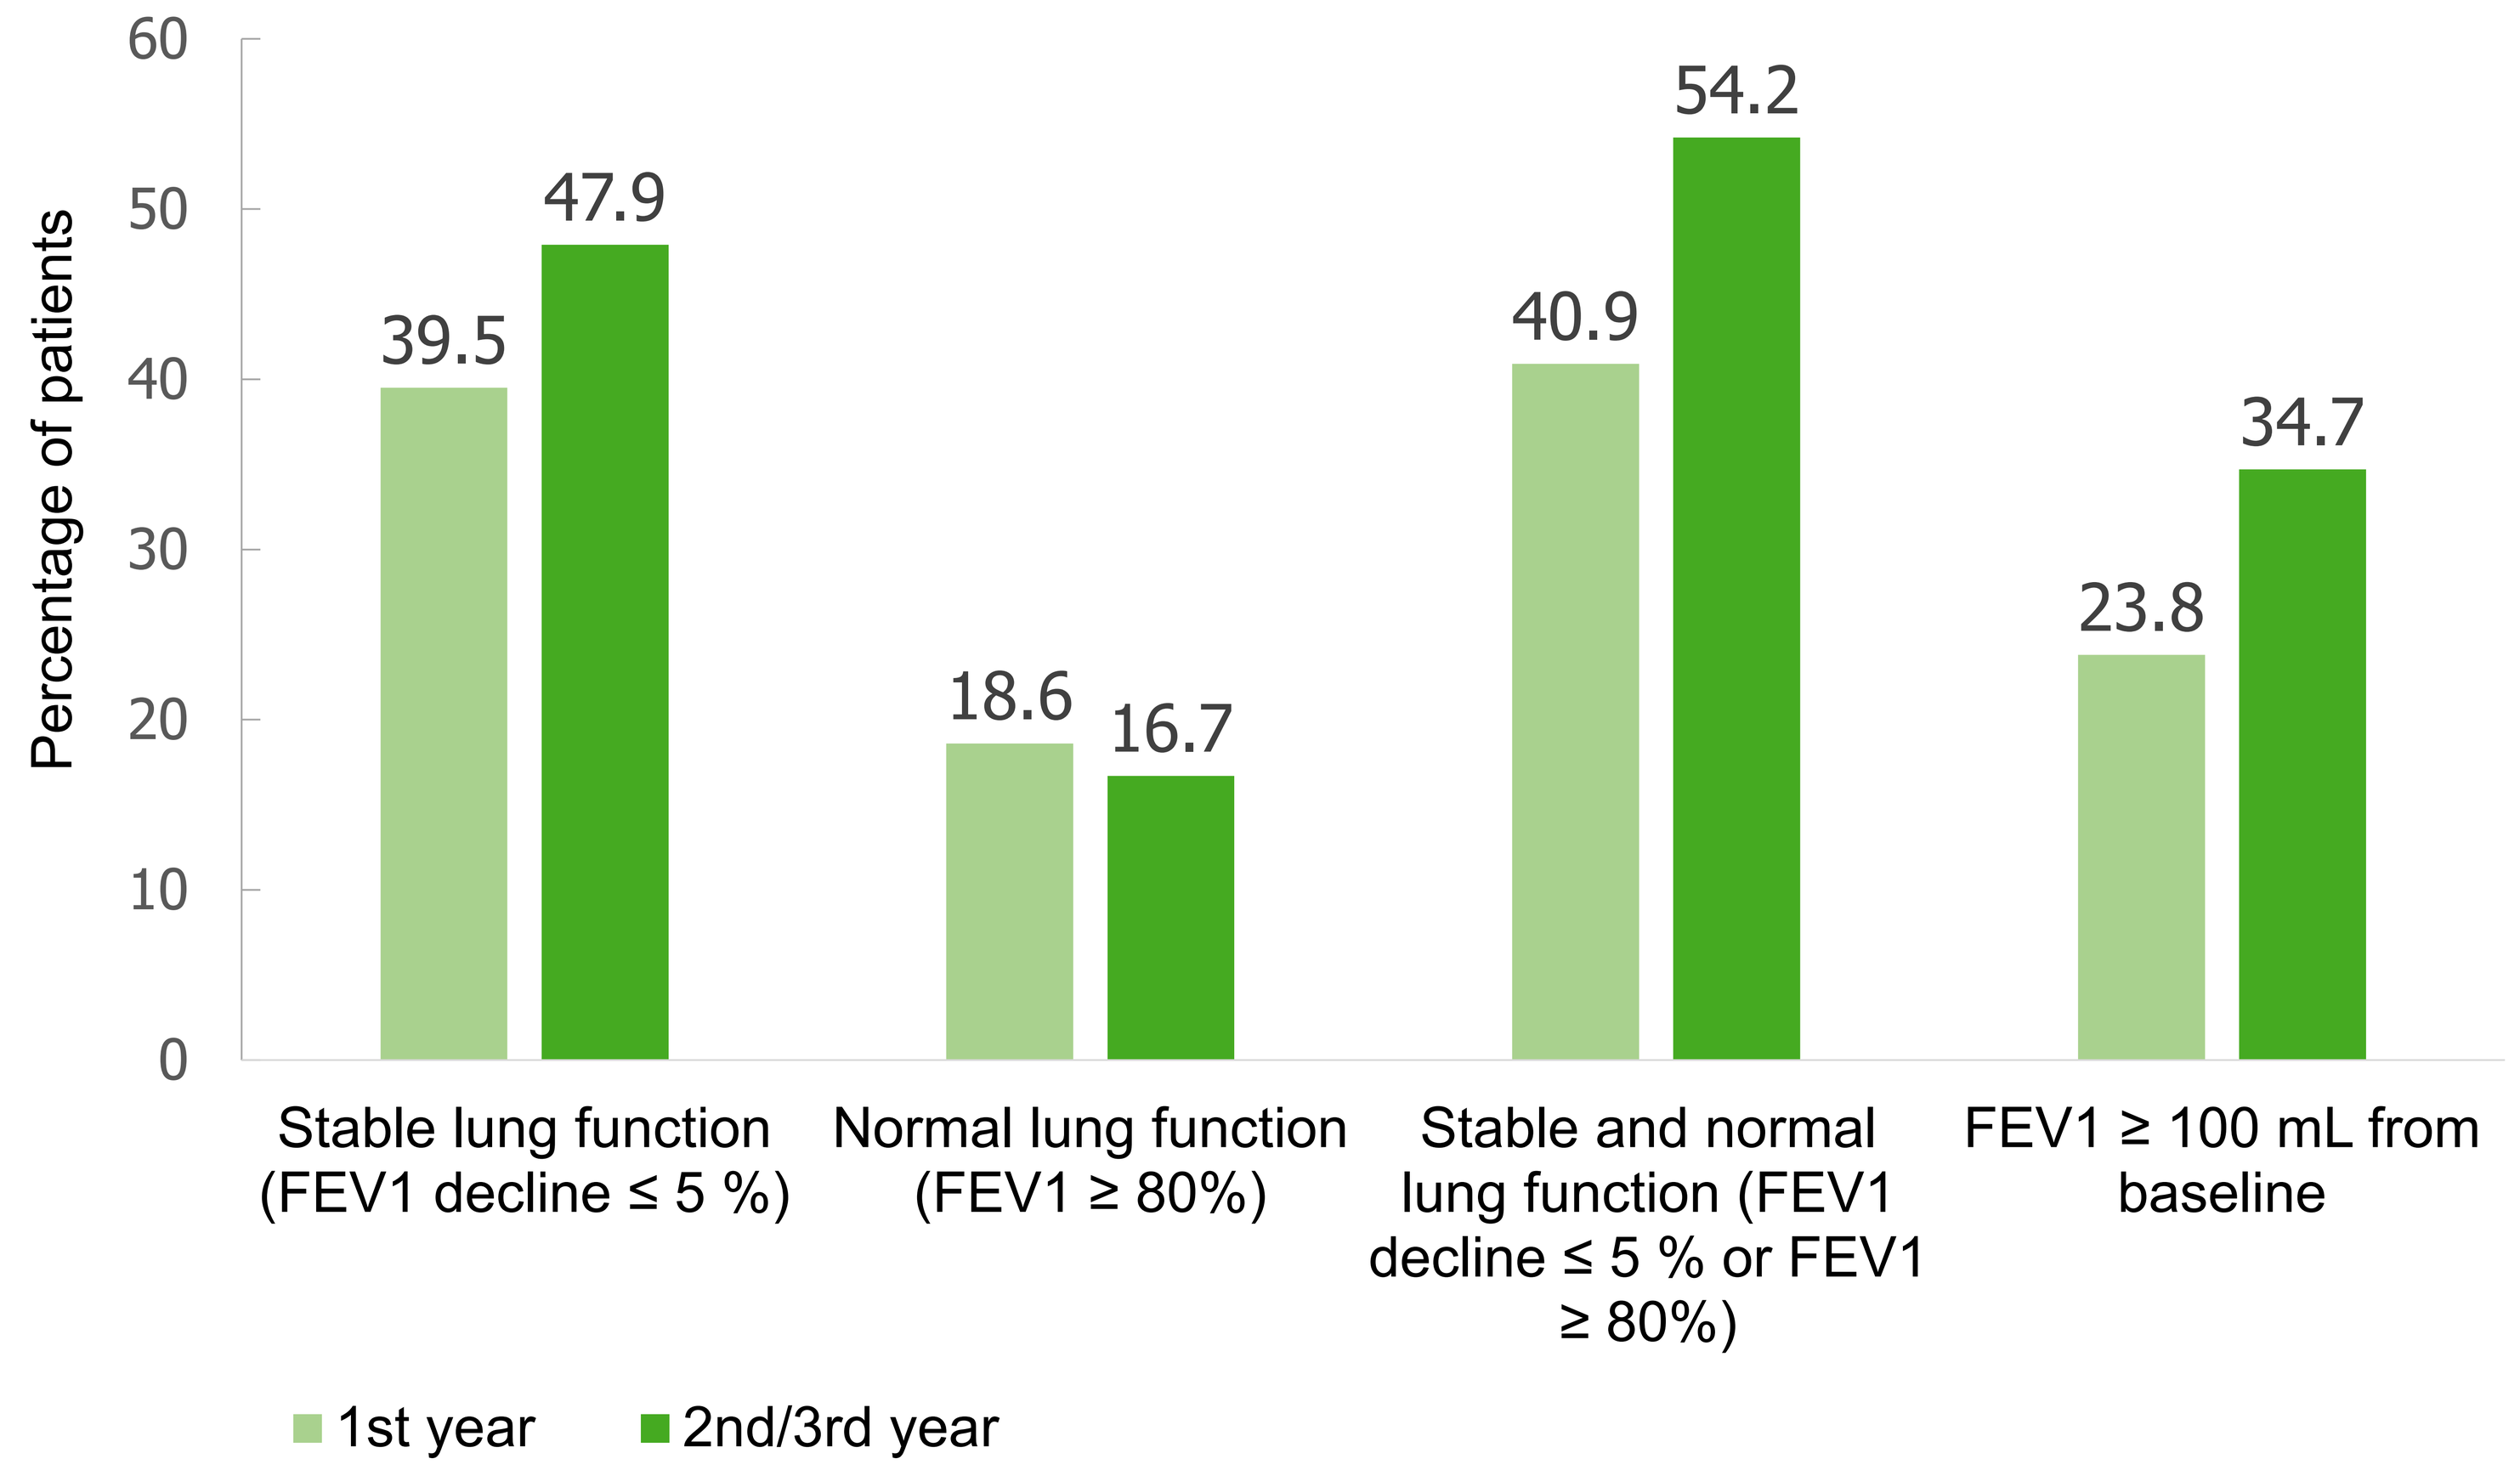
Percentage of patients
60
50
40
30
20
10
0
39.5
47.9
18.6
16.7
40.9
54.2
23.8
34.7
Stable lung function (FEV1 decline ≤ 5 %)
Normal lung function (FEV1 ≥ 80%)
Stable and normal lung function (FEV1 decline ≤ 5 % or FEV1 ≥ 80%)
FEV1 ≥ 100 mL from baseline
1st year
2nd/3rd year

Among different lung function criteria, stable lung function appears to best reflect clinical remission in real-world practice. In contrast, normal lung function and absolute FEV_1 thresholds identify fewer patients and show less persistence across follow-up.

Figure 4. Sankey diagram showing transitions across clinical remission categories

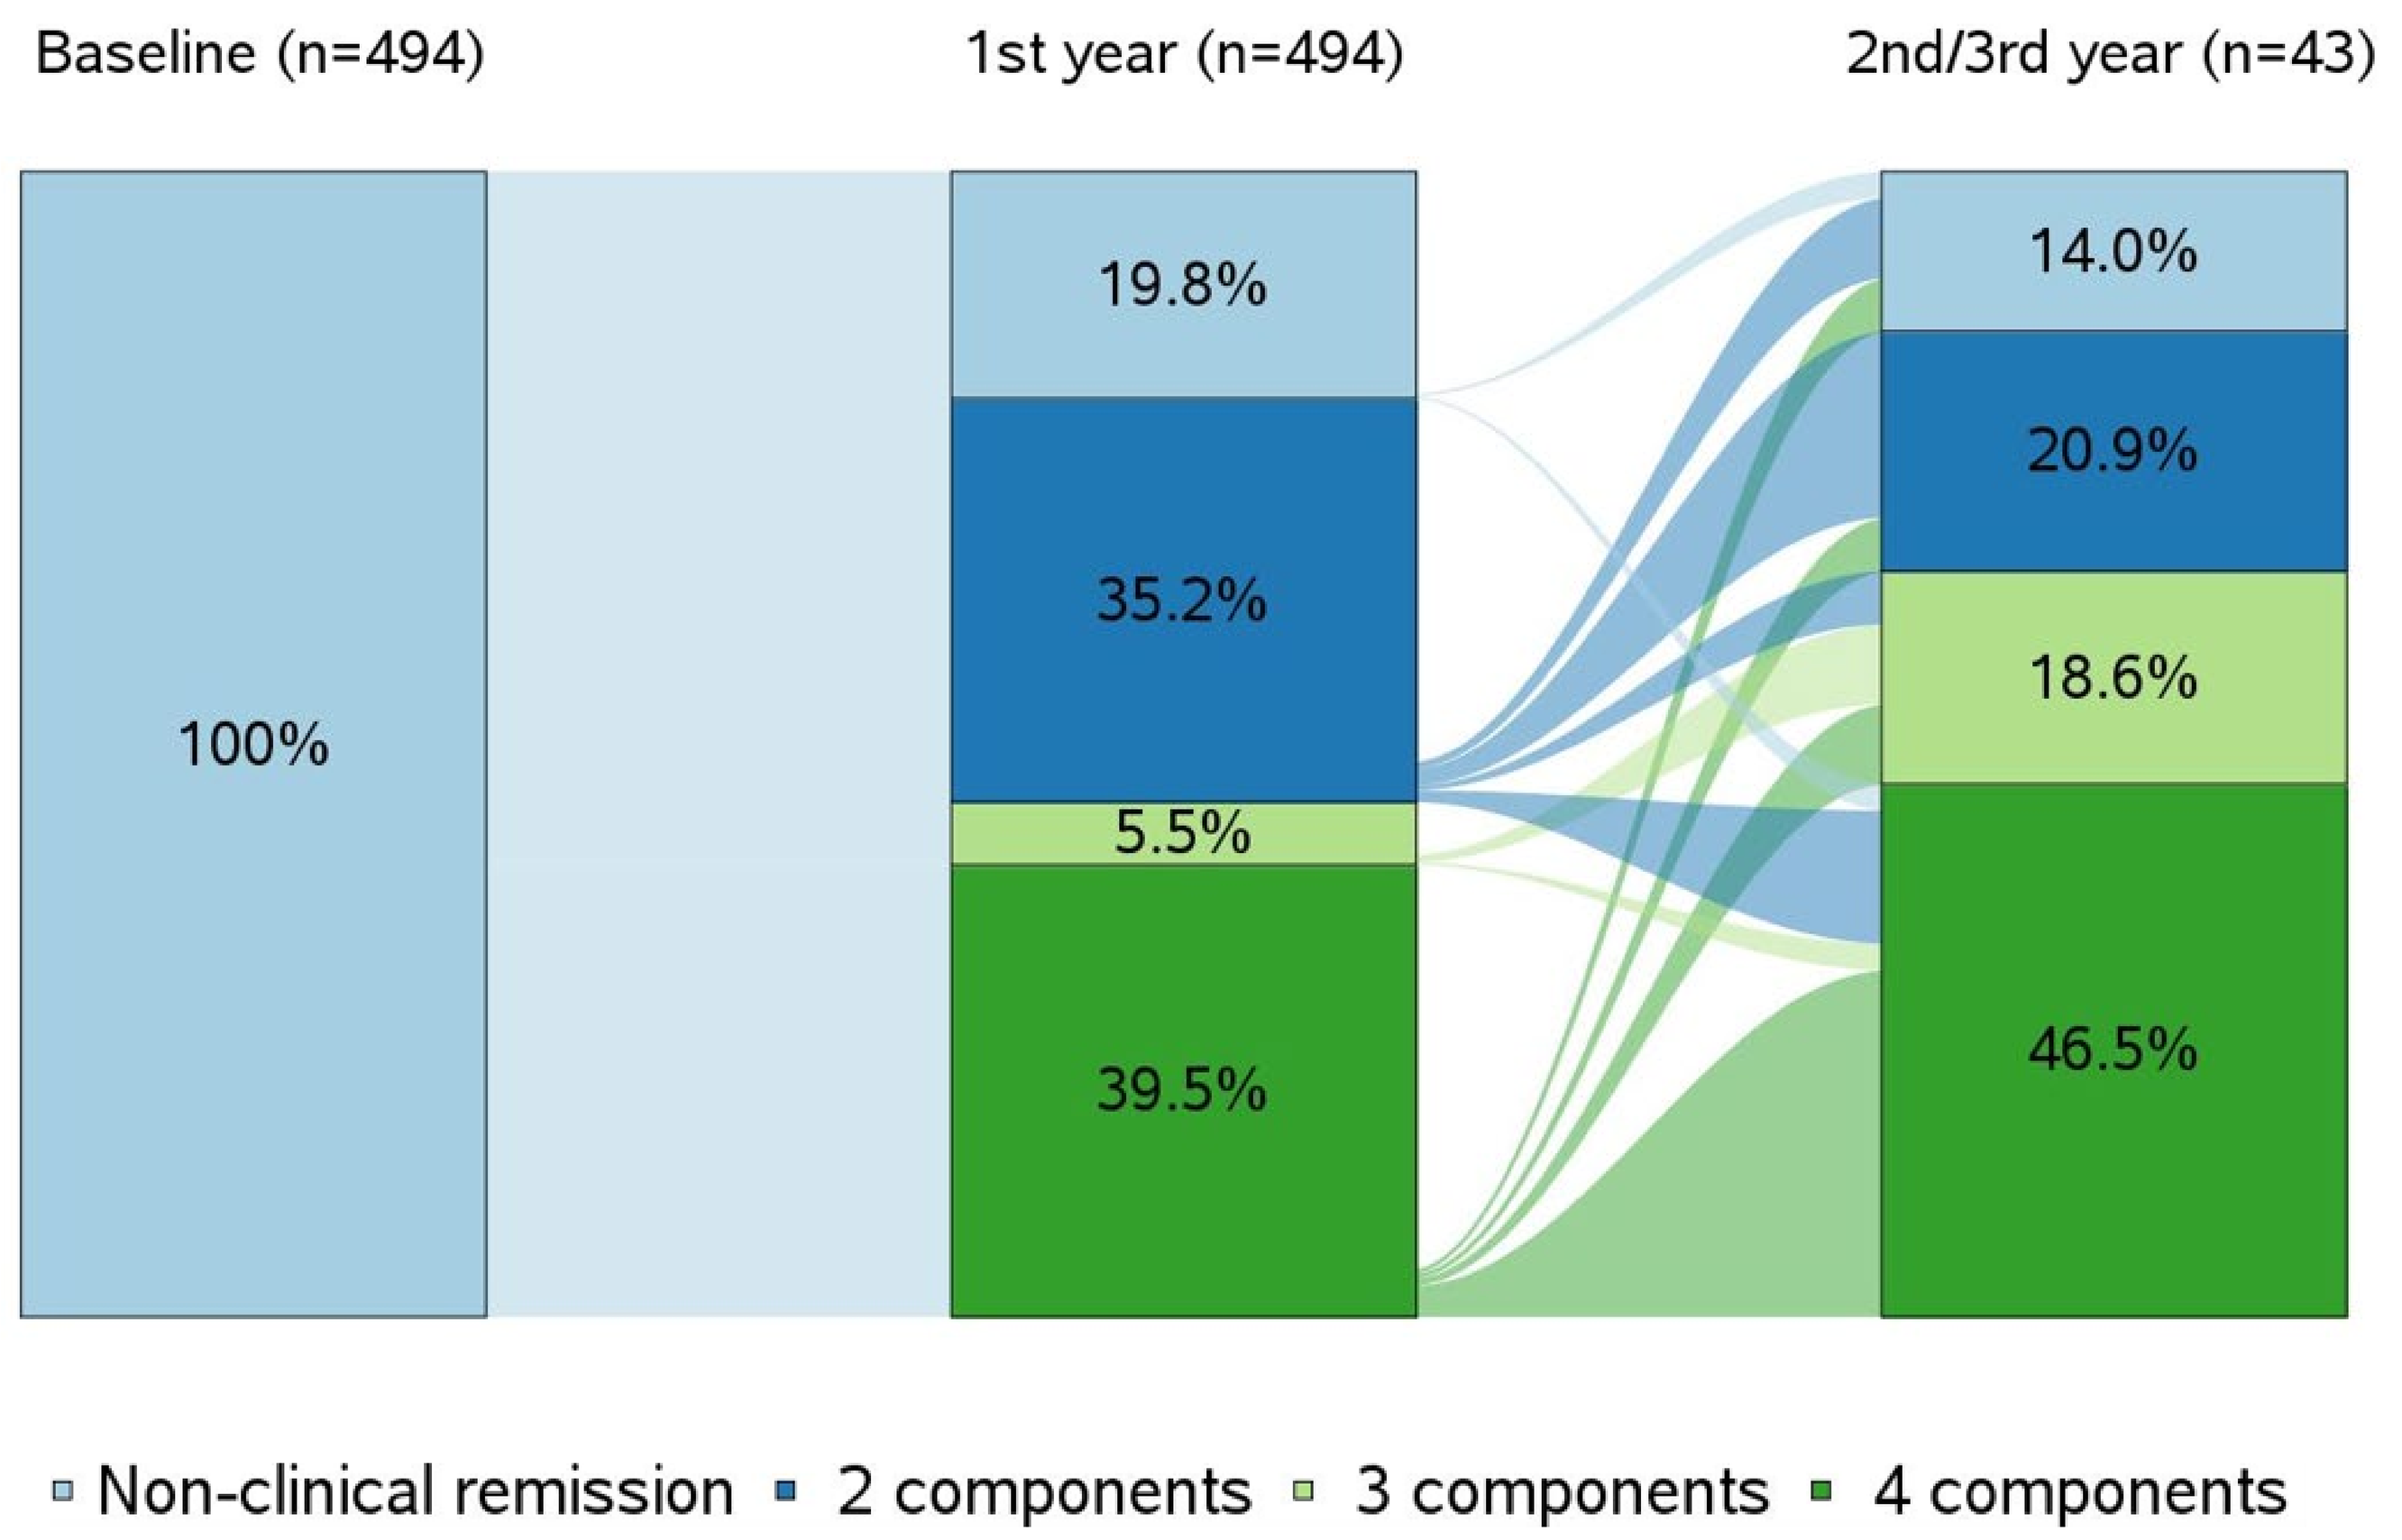
Baseline (n=494)
1st year (n=494)
2nd/3rd year (n=43)
100%
19.8%
35.2%
5.5%
39.5%
14.0%
20.9%
18.6%
46.5%
Non-clinical remission
2 components
3 components
4 components

CONCLUSIONS:

Clinical remission can be achieved in a large proportion of patients after 1 year of BDP/FF/G treatment in a real-world setting, and this proportion further increases in the following years.

References:

[1]Virchow J.C. et al., Single inhaler extrafine triple therapy in uncontrolled asthma (TRIMARAN and TRIGGER): two double-blind, parallel-group, randomised, controlled phase 3 trials. The Lancet, 2019. 394(10210): p. 1737-1749.

[2]Gessner C, et al. Real-World Effectiveness and Safety of Single Inhaler Triple Therapy with Beclometasone/ Formoterol/ Glycopyrronium in Moderate to Severe Asthma: TriMaximize Study. J Asthma Allergy. 2026. doi:10.2147/JAA.S582286

The TriMaximize study was funded by Chiesi. CG, FT, REKR, CSU, WP, AB, VP, MK and FB have received fees for conducting the study. VB, CF, VG and AH are employees of Chiesi during the planning, implementation or evaluation of the study.